Pharmacokinetic Results of Single-Dose and Multiple-Dose Bioequivalence Studies of Milsaperidone and Iloperidone Immediate-Release Oral Tablets

Sean R. Chadwick PhD†, Rosarelis Torres PhD†, Changfu Xiao PhD†, Christos M. Polymeropoulos MD†, Gunther Birznieks MS†, Mihael H. Polymeropoulos MD†
† *Vanda Pharmaceuticals, Inc., Washington, DC 20037*
American Society For Clinical Psychopharmacology, 2025 Annual Meeting, Scottsdale, AZ 85255

Aim

Evaluate the pharmacokinetics and comparative bioavailability of milsaperidone compared to iloperidone

Background

Milsaperidone (formerly VHX-896 and P88) is the major active metabolite of iloperidone. Milsaperidone crosses the blood brain barrier, where exposure to the metabolite is believed to contribute to the clinical effects of iloperidone[1].

Methods

Single-dose study design:

- 3mg single oral fasted comparison in healthy volunteers
- Bioequivalence: LSGMR estimates, 90% CIs range 80%-125%[3]
- PK parameters (plasma): C_{max}, $AUC_{0\text{-}t}$, $AUC_{0\text{-}inf}$

Screening	Period 1		Period 2
A (TEST-REF)	**3mg milsaperidone** (three 1mg tablets)	→	**3mg iloperidone** (three 1mg tablets)
OR	**Treatment #1**	**Crossover**	**Treatment #2**
B (REF-TEST)	**3mg iloperidone** (three 1mg tablets)	→	**3mg milsaperidone** (three 1mg tablets)
Randomized to treatment sequence at baseline visit	**PK testing** (5 days)	**Wash-Out** (14 days)	**PK testing** (5 days)

Multiple-dose study design:

- 12mg repeat oral bid dose fasted comparison
- Patients with schizophrenia or bipolar I disorder
- Bioequivalence: LSGMR estimates, 90% CIs range 80%-125%[3]
- PK parameters (plasma): C_{max} and $AUC_{0\text{-}12\ hours}$

	Treatment Period 1		Treatment Period 2
Screening (Day ≥ -1 CYP2D6 Results ✓)	**Treatment 1** *Up Titration (6 days)*	*Maintenance (9 days)*	**Treatment 2** *Maintenance (9 days)*
Randomization Sequence		Crossover	
A (TEST-REF)	Milsaperidone 1mg→10mg twice daily	Milsaperidone 12mg twice daily	Iloperidone 12mg twice daily
OR		*NO Washout*	
B (REF-TEST)	Iloperidone 1mg→10mg twice daily	Iloperidone 12mg twice daily	Milsaperidone 12mg twice

Day 0	Day 1	Day 6	Day 7 to 14	Day 15	Day 16	Day 17 to 23	Day 24
Baseline Visit Randomization	Treatment Starts	Titration Complete	Maintenance Trough PK	PK $AUC_{0\text{-}12}$	PK $AUC_{0\text{-}12}$	Maintenance Trough PK	PK $AUC_{0\text{-}12}$

Receptor pharmacology

Milsaperidone possesses a similar binding profile to the parent drug, with a strong binding affinity for adrenergic alpha (NEα1 and α2c), serotonin (5HT2a), and dopamine (D2) receptors[1,2]

Milsaperidone and iloperidone are bioequivalent following single oral doses

Iloperidone
(geometric mean concentration)

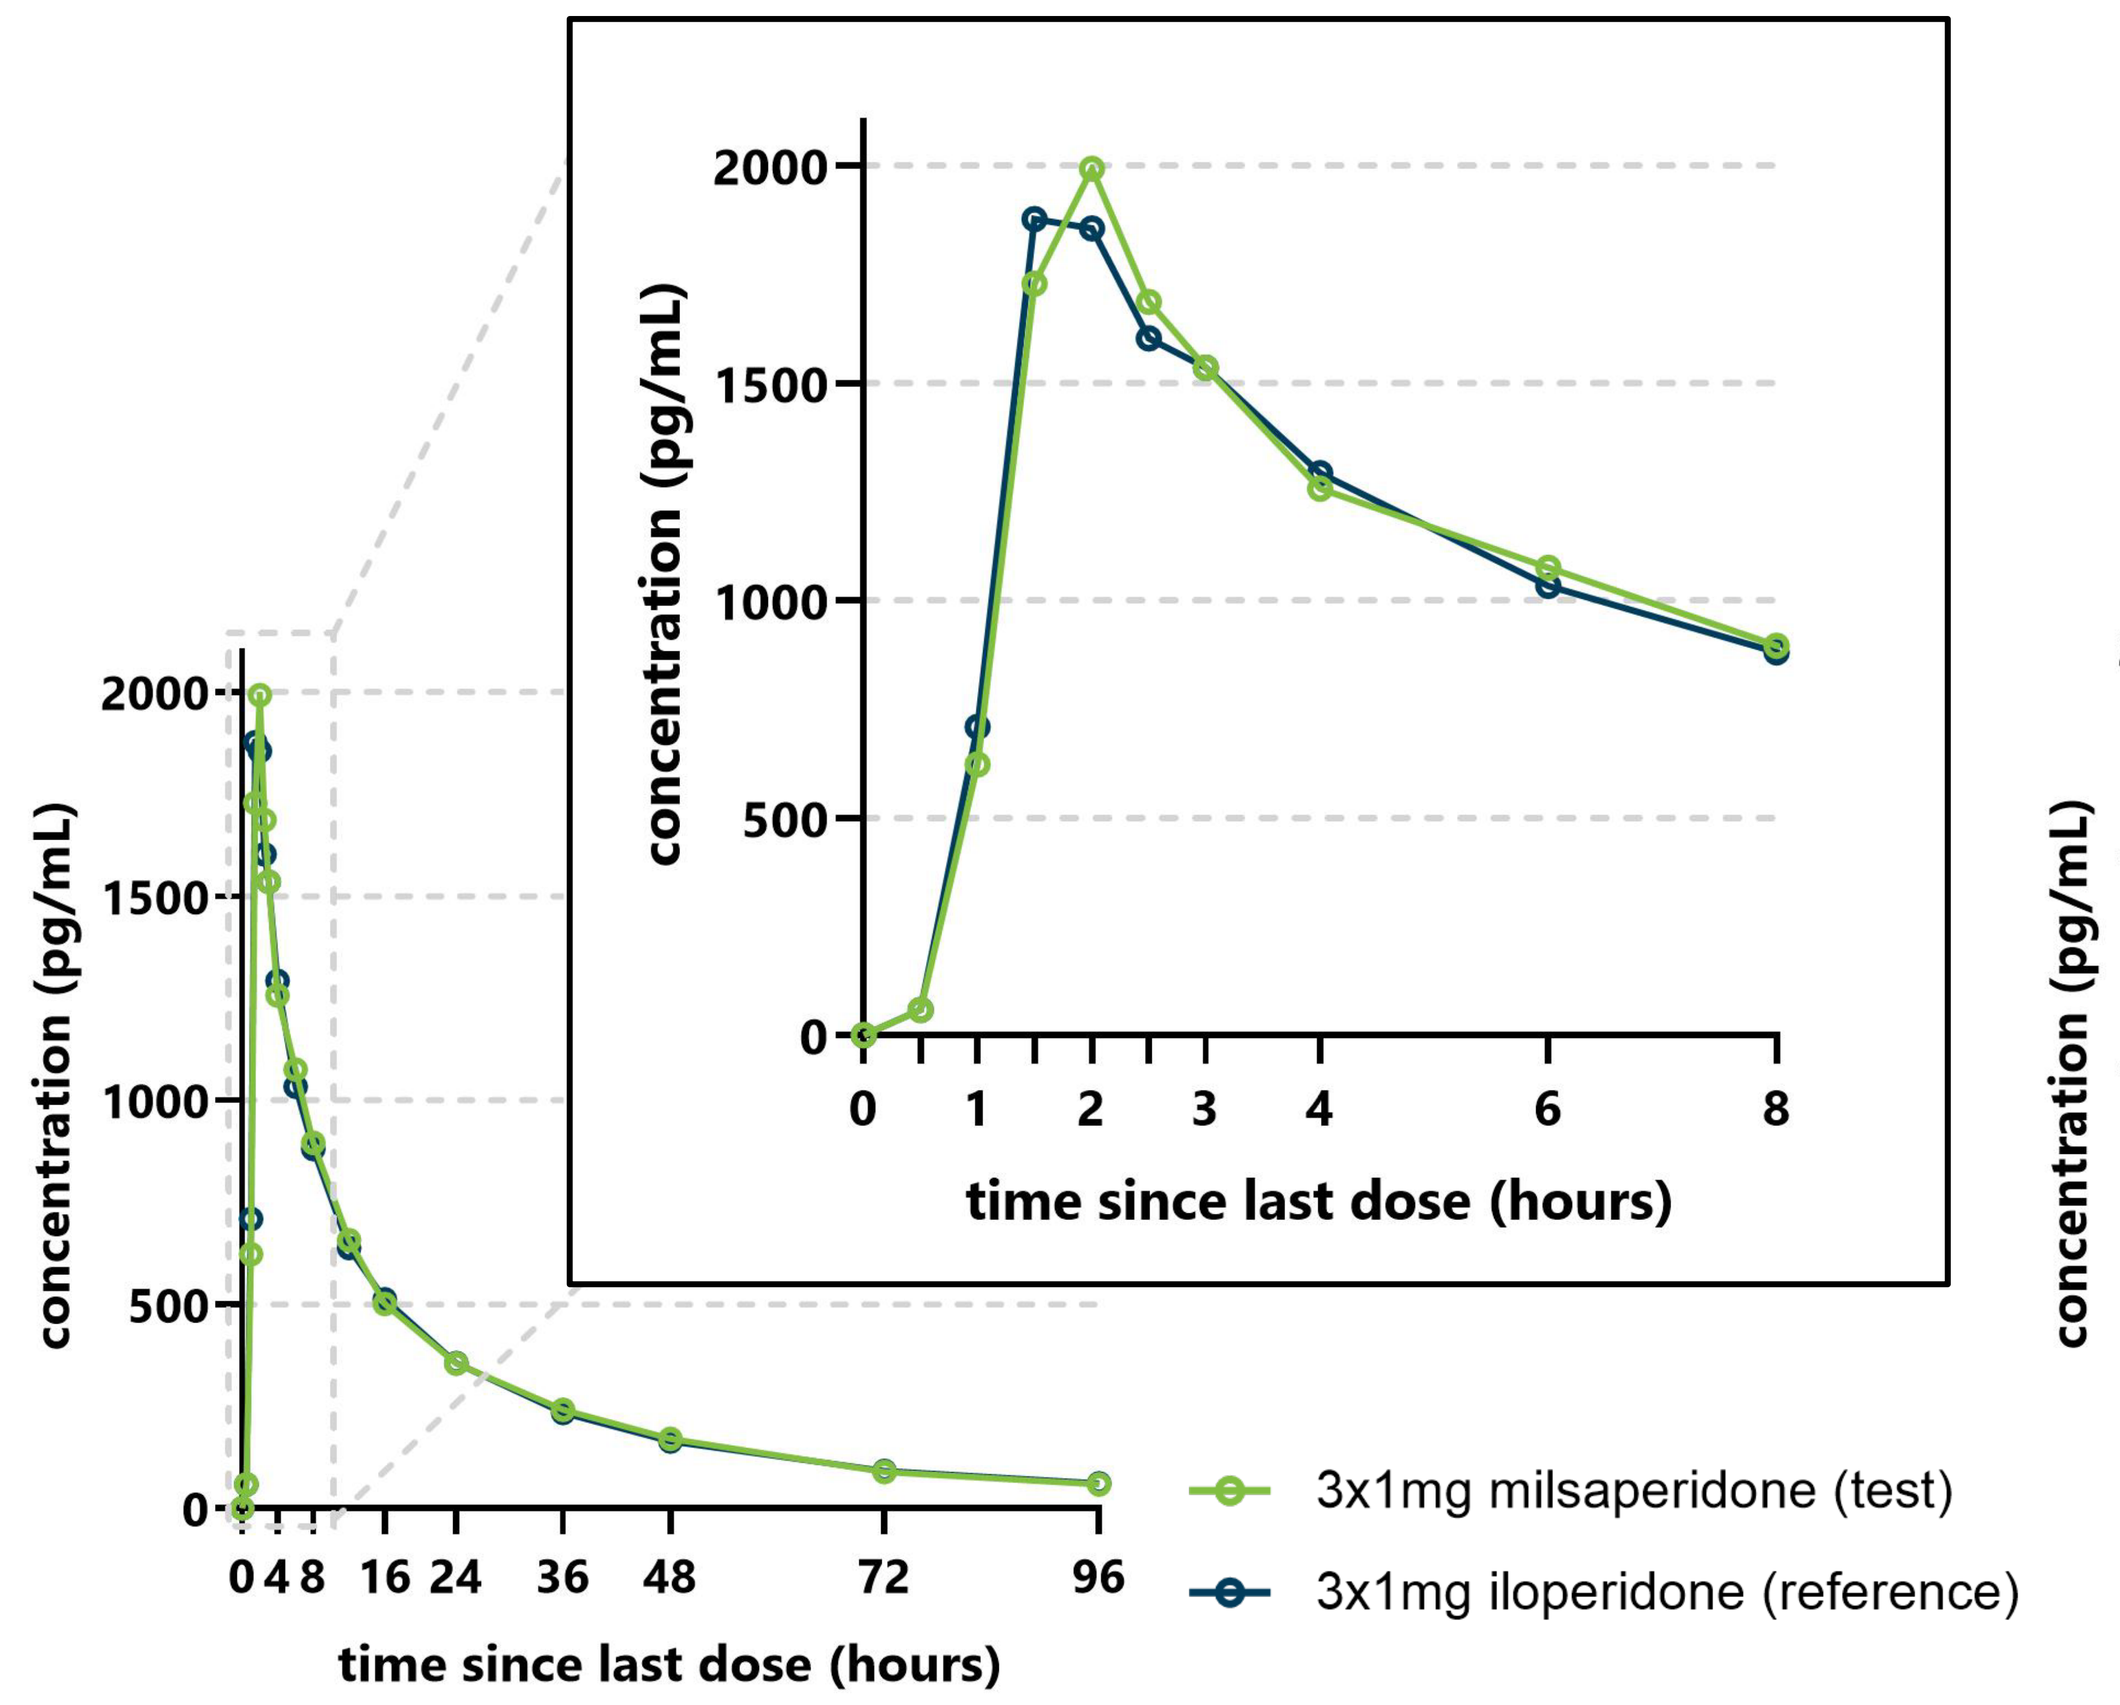
concentration (pg/mL)
time since last dose (hours)
3x1mg milsaperidone (test)
3x1mg iloperidone (reference)

Milsaperidone
(geometric mean concentration)

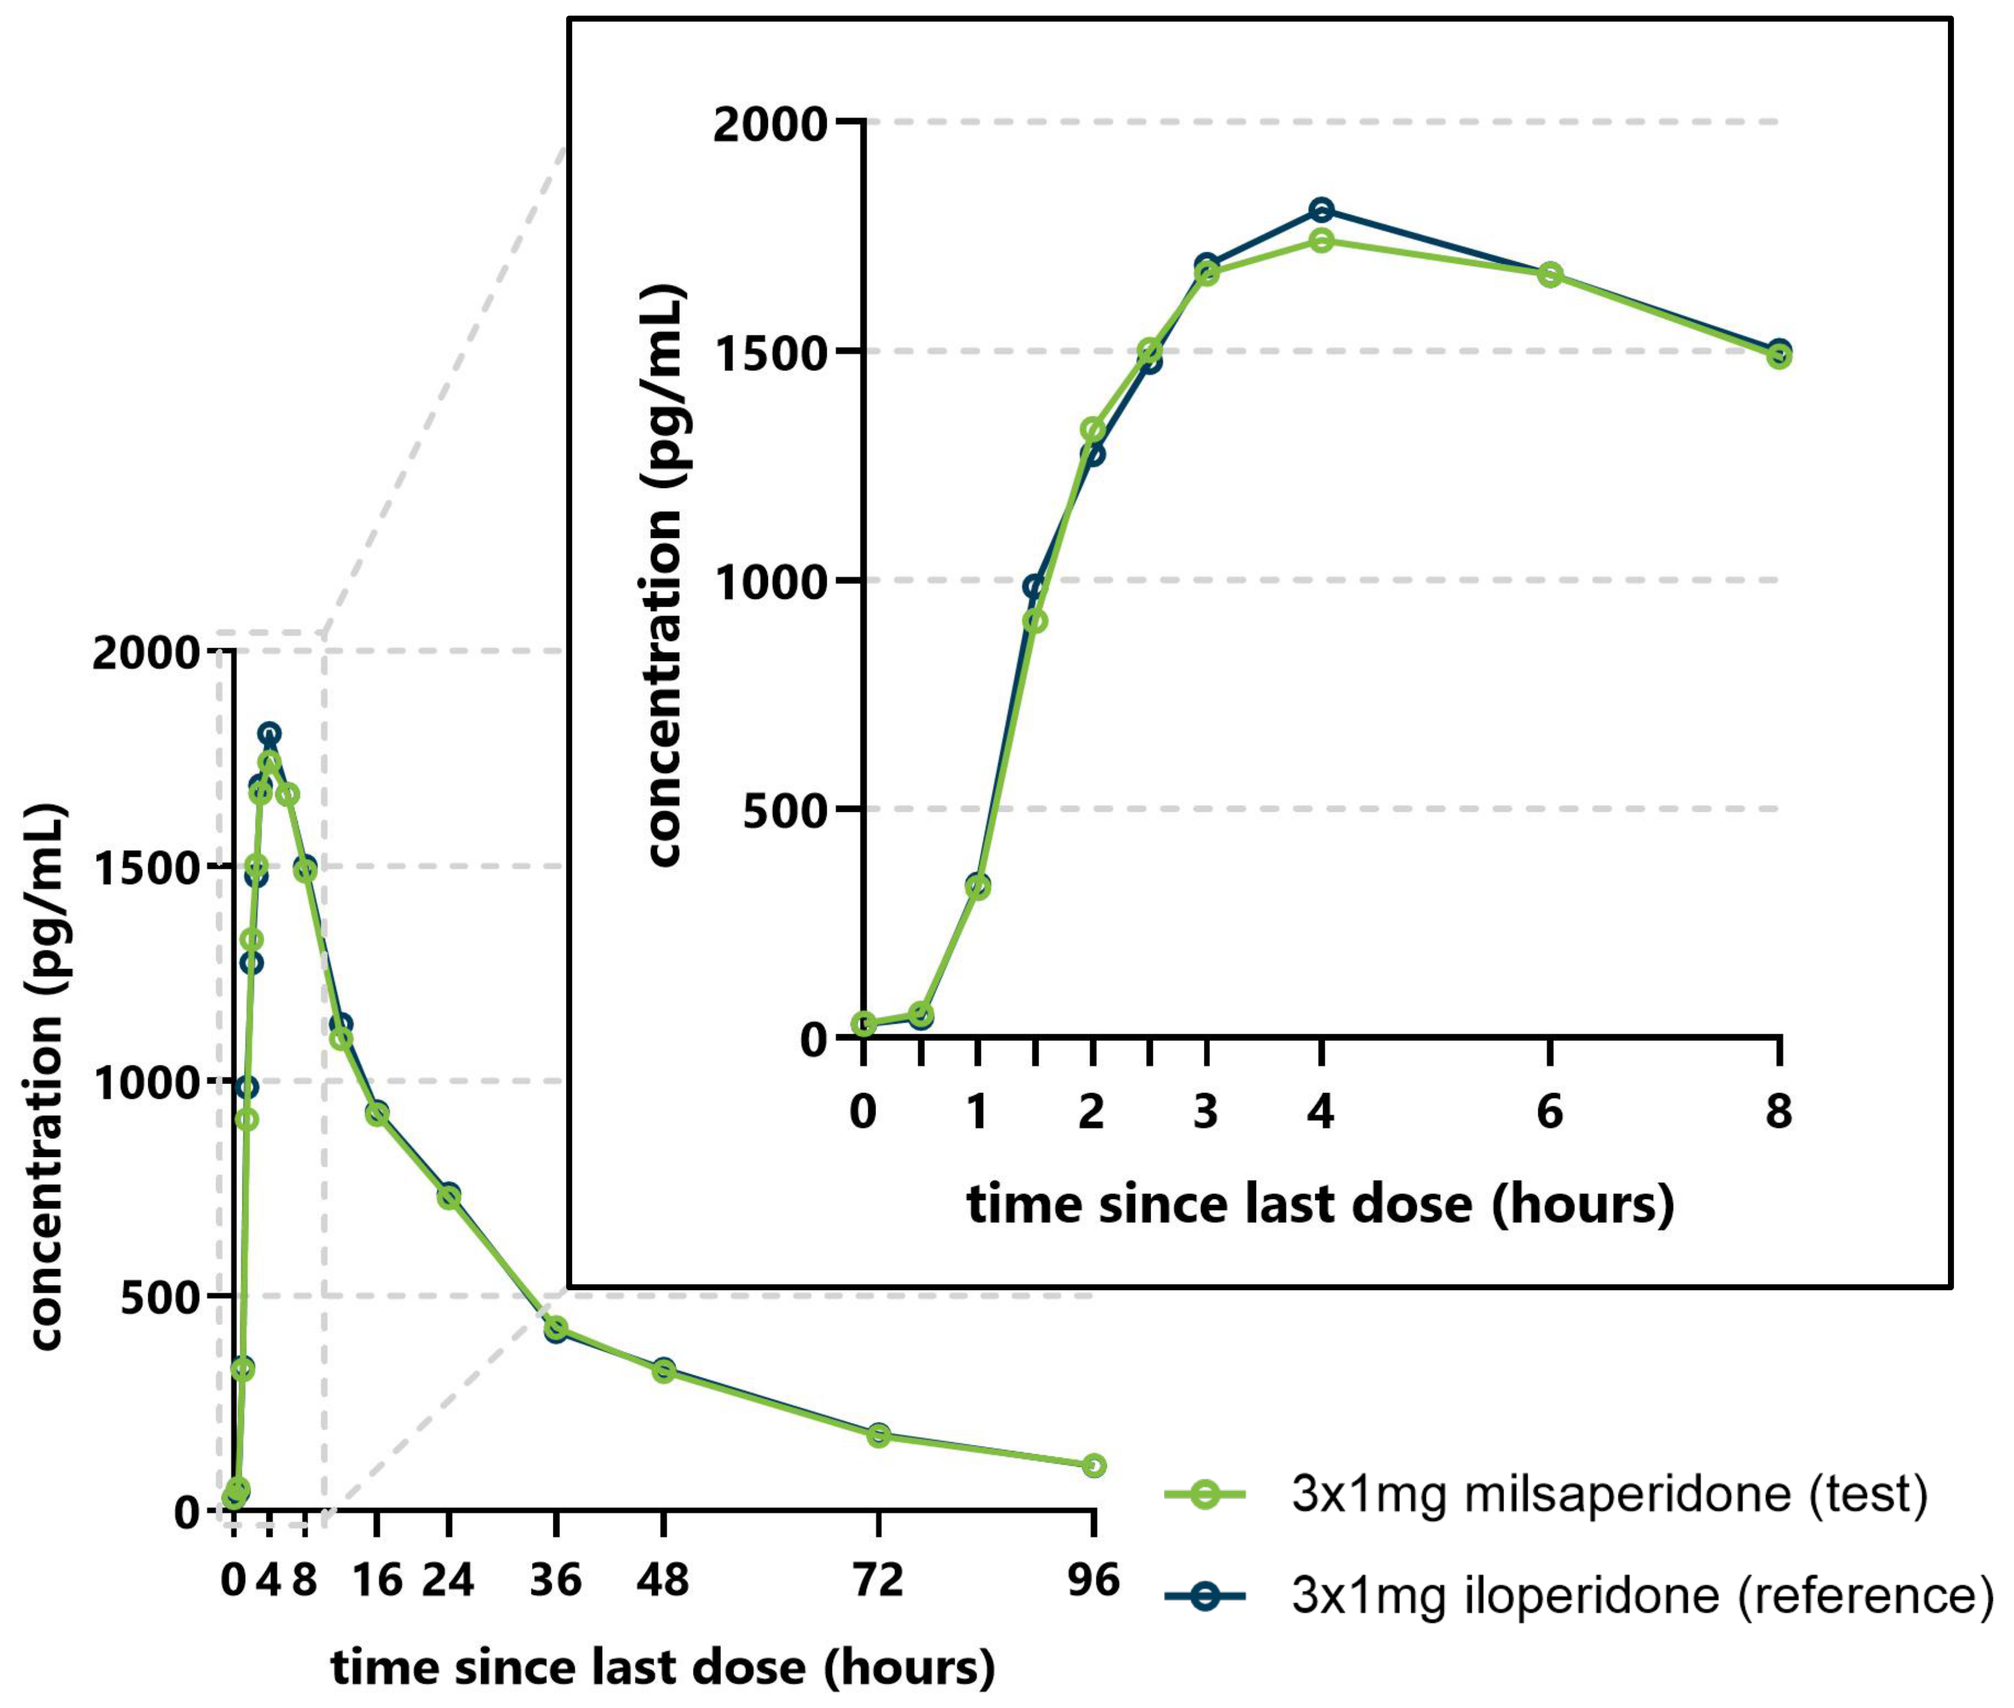
concentration (pg/mL)
time since last dose (hours)
3x1mg milsaperidone (test)
3x1mg iloperidone (reference)

Milsaperidone and iloperidone are bioequivalent at steady state

Iloperidone
(geometric mean concentration)

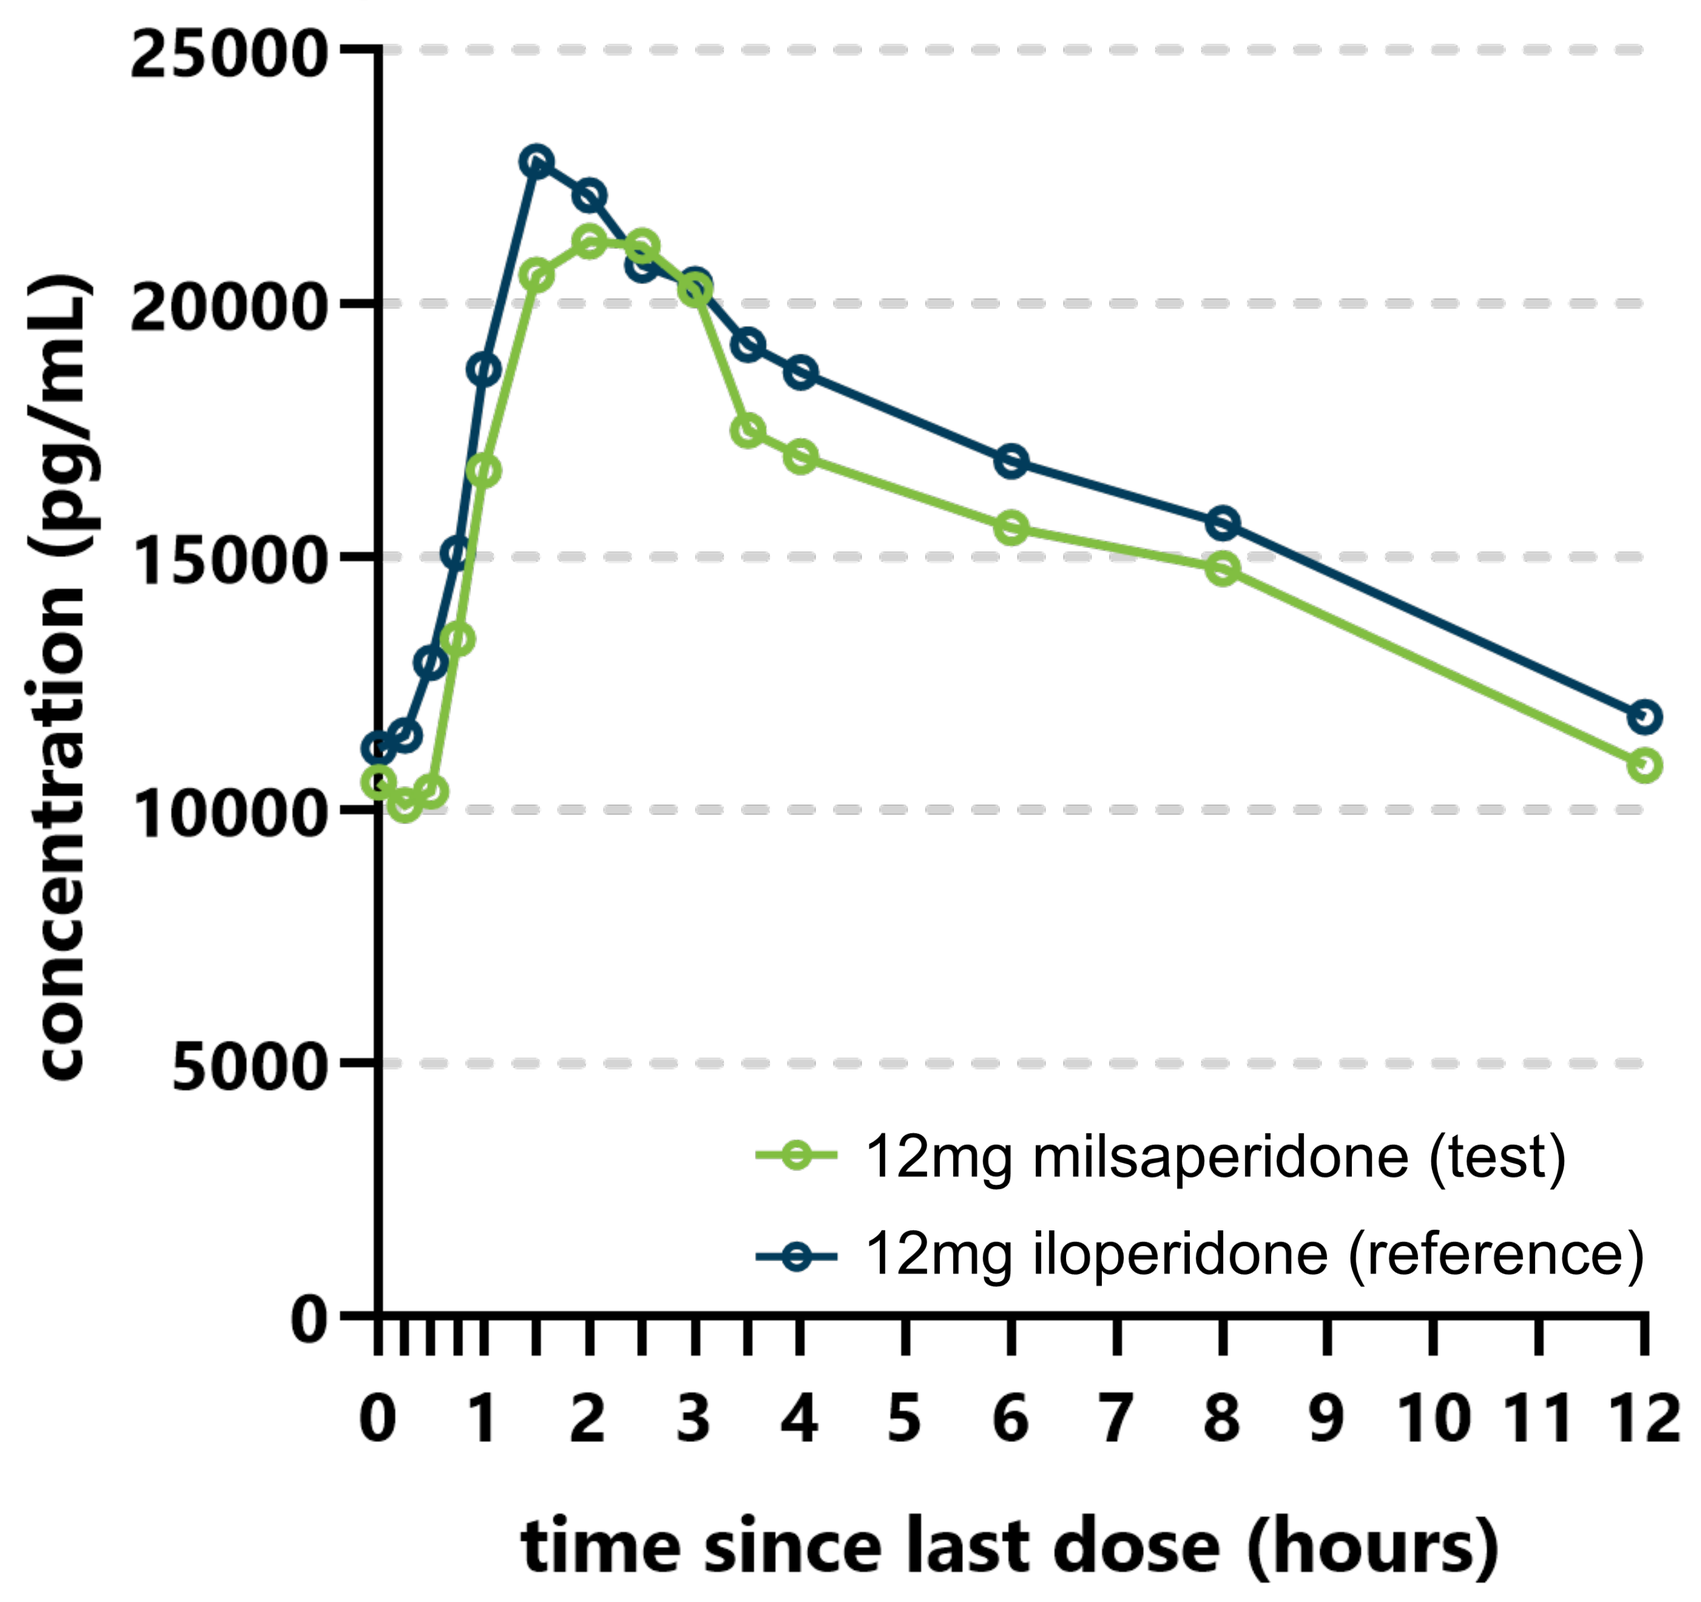
concentration (pg/mL)
time since last dose (hours)
12mg milsaperidone (test)
12mg iloperidone (reference)

Milsaperidone
(geometric mean concentration)

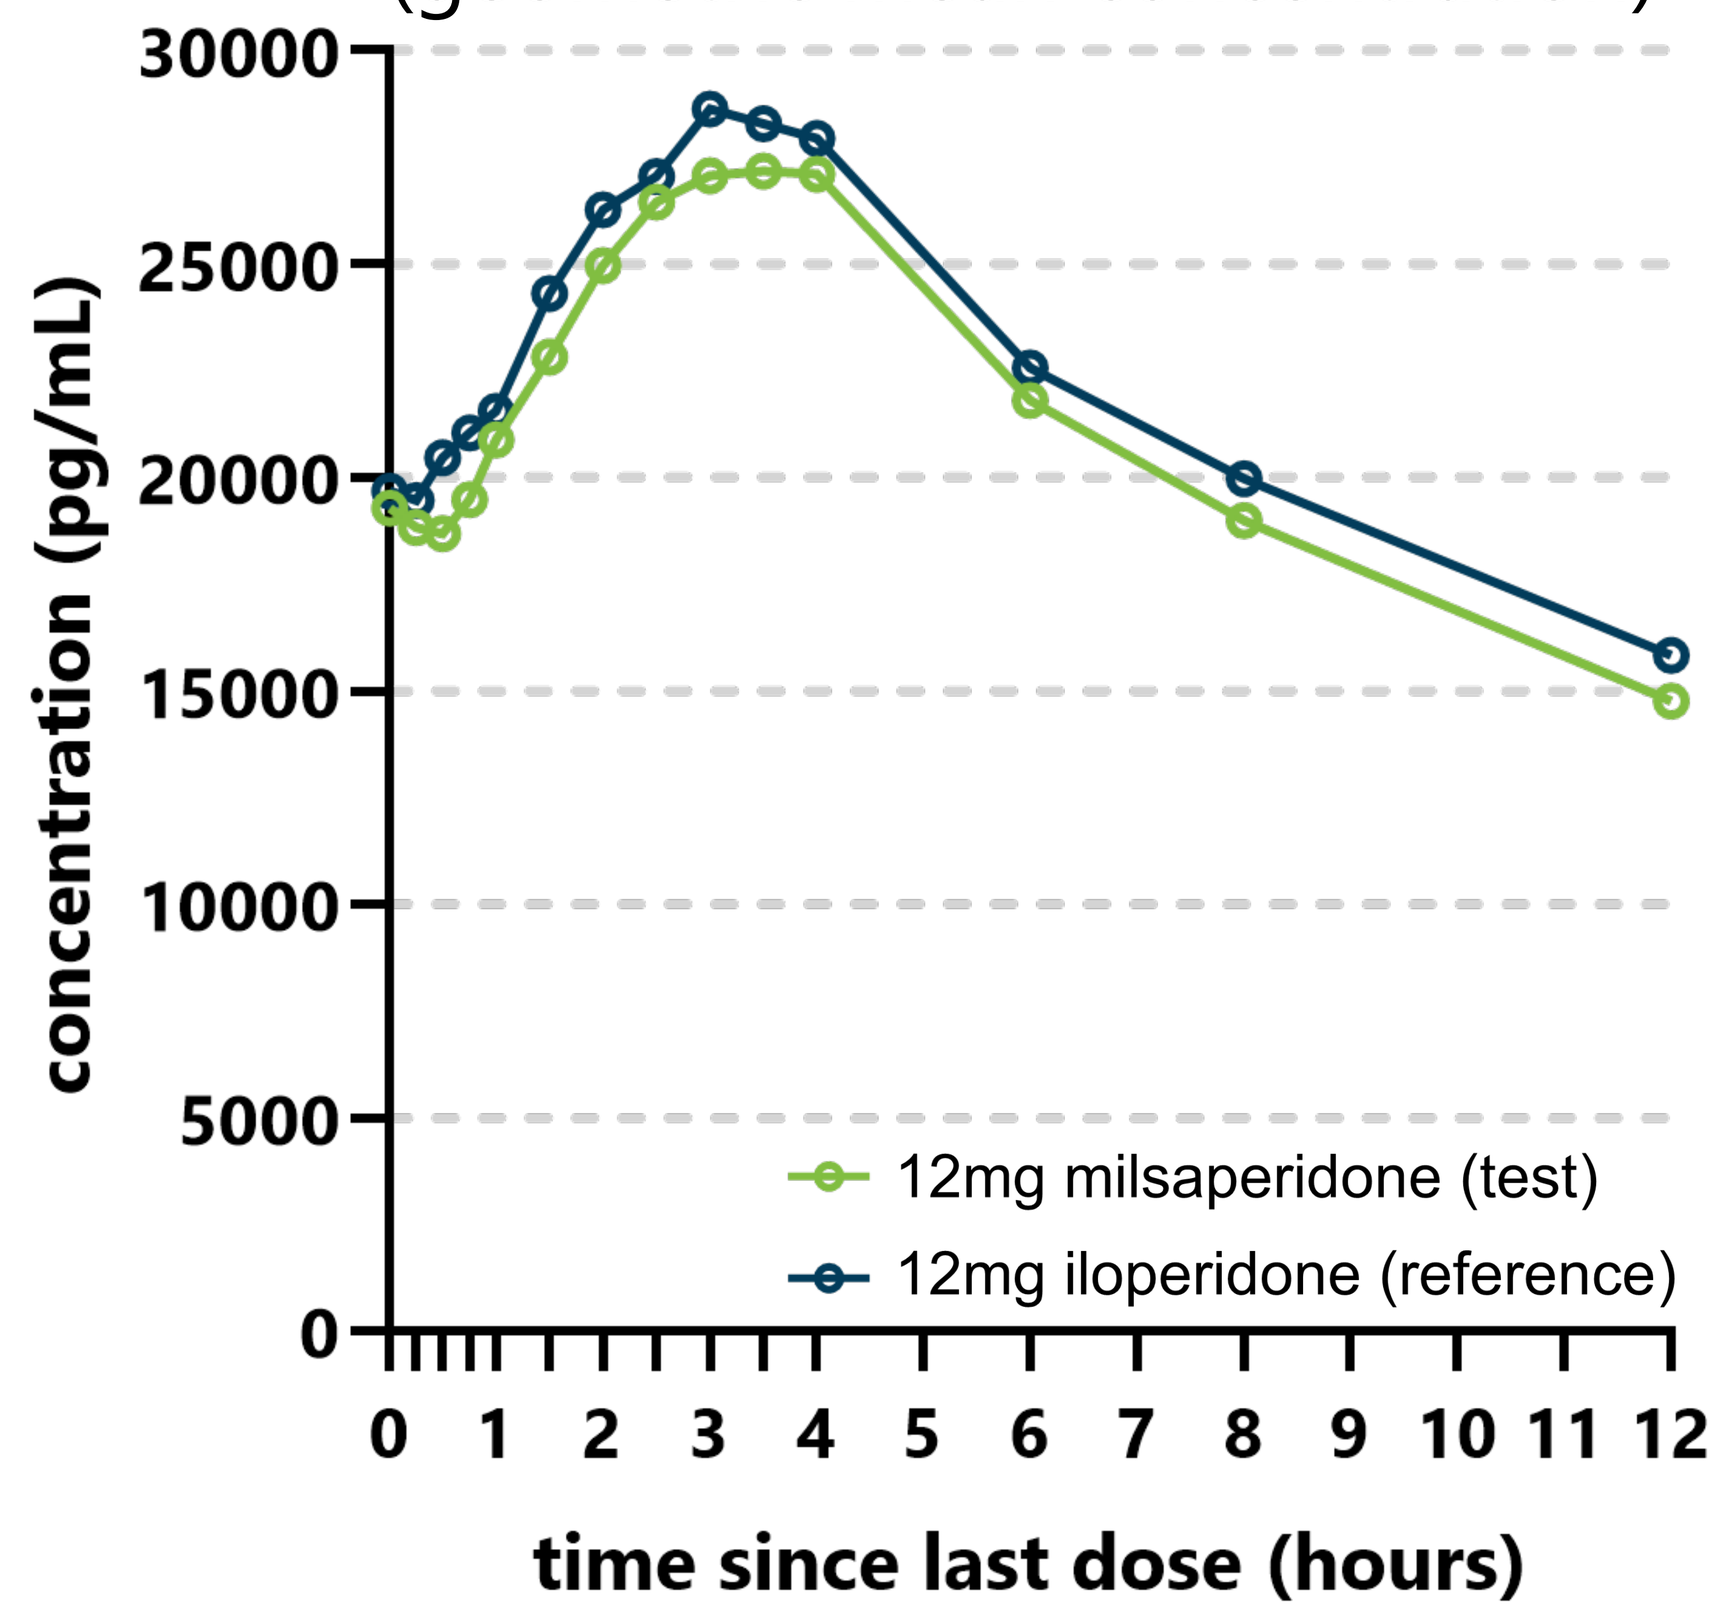
concentration (pg/mL)
time since last dose (hours)
12mg milsaperidone (test)
12mg iloperidone (reference)

Single-dose pharmacokinetic results

Iloperidone pharmacokinetic parameters and estimates

Parameter N=19	Milsaperidone 3x1mg (test)	Iloperidone 3x1mg (reference)	GMR or estimate (90% CI)
Cmax, pg/mL	**2,309**	**2,257**	**102.33** (94.00–111.40)*
AUC(0-t), hr×pg/mL	**29,813**	**29,648**	**100.56** (95.97–105.36)*
AUC(inf), hr×pg/mL	**32,511**	**32,575**	**99.80** (95.23–104.60)*
Tmax, hours (range)	**2.0** (1.0 – 3.0)	**1.5** (1.0 – 4.0)	.

Milsaperidone pharmacokinetic parameters and estimates

Parameter N=19	Milsaperidone 3x1mg (test)	Iloperidone 3x1mg (reference)	GMR or estimate (90% CI)
Cmax, pg/mL	**1,845**	**1,881**	**98.08** (91.00–105.71)*
AUC(0-t), hr×pg/mL	**47,961**	**48,655**	**98.58** (94.08–103.29)*
AUC(inf), hr×pg/mL	**52,323**	**53,267**	**98.23** (93.97–102.67)*
Tmax, hours (range)	**4.0** (2.0 – 8.0)	**4.0** (3.0 – 8.0)	.

* Estimate(s) and 90% CIs were within limits of 80-125% per FDA guidance for determining bioequivalence[3]

Steady-state pharmacokinetic results

Iloperidone pharmacokinetic parameters and estimates

Parameter N=20	Milsaperidone 12mg (test)	Iloperidone 12mg (reference)	GMR or estimate (90% CI)
Cmax, pg/mL	**24,840**	**25,610**	**97.00** (94.70–99.35)*
AUC(0-12), hr×pg/mL	**186,633**	**200,856**	**92.92** (91.83–94.02)*
Tmax, hours (range)	**2.0** (1.0 – 8.0)	**2.0** (1.0 – 6.0)	.

Milsaperidone pharmacokinetic parameters and estimates

Parameter N=20	Milsaperidone 12mg (test)	Iloperidone 12mg (reference)	GMR or estimate (90% CI)
Cmax, pg/mL	**28,889**	**30,365**	**95.14** (93.93–96.37)*
AUC(0-12), hr×pg/mL	**253,310**	**265,887**	**95.27** (94.08–96.48)*
Tmax, hours (range)	**3.5** (2.0 – 4.1)	**3.5** (2.0 – 4.1)	.

* Estimate(s) and 90% CIs were within limits of 80-125% per FDA guidance for determining bioequivalence[3]

Conclusions

Milsaperidone and iloperidone interconvert and are bioequivalent across the entire approved[2] therapeutic dose range of iloperidone.

Future Development

Milsaperidone is currently under phase 3 evaluation for treatment in major depressive disorder adjunctive to antidepressant therapy in adults (NCT06830044)

Footnotes: Single dose study: NCT06494397; Repeat dose Study: NCT04969211. Cmax and AUC parameters report least squares geometric means rounded to the nearest pg/mL or hxp/mL, respectively. Tmax is reported as median (range), rounded to the nearest 0.1 hours. $AUC_{0\text{-}t}$ was calculated according to the linear trapezoidal rule where t is the last observable concentration above the limit of quantification, and AUC_{inf} was extrapolated to infinity by dividing the last measured concentration by the terminal elimination rate constant. GMRs reflect milsaperidone/iloperidone (i.e., test/reference) expressed as a percentage.

Abbreviations: AUC, area under the plasma concentration-time curve; bid, twice daily; CI, confidence interval; Cmax, maximum concentration; CYP2D6, Cytochrome P450 2D6 enzyme, LSGMR, least squares geometric mean ratio; hr, hour; inf, infinity; pg/mL, picograms per milliliter; PK, pharmacokinetic; mg, milligram; t, time; ref, reference study medication; Tmax, time of peak concentration.

References: 1. Subramanian & Kalkman, (2002). *Progress in Neuro-Psychopharmacology and Biological Psychiatry*. DOI: https://doi.org/10.1016/S0278-5846(01)00307-4. 2. Vanda Pharmaceuticals Inc. (2025). *Fanapt (iloperidone) USPI*. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2025/022192s024lbl.pdf. Accessed 22 May 2025. 3. US Food and Drug Administration (FDA), (2021), *Guidance for Industry: Bioequivalence Studies With Pharmacokinetic Endpoints*. Available at: https://www.fda.gov/media/87219/download. Accessed 22 May 2025.

VANDA PHARMACEUTICALS INC.